Consistent Outcomes With Canagliflozin in Patients With Type 2 Diabetes Across Geographic Regions: Results From the CANVAS Program

CANVAS Program

Carol Wysham,[1,*] Arpandev Bhattacharyya,[2] Michael Tsoukas,[3] Esperanza Martínez-Abundis,[4] April Slee,[5] Wayne Shaw,[6] Kenneth W. Mahaffey,[7] Dick de Zeeuw[8]

[1]Washington State University, Spokane, WA, USA; [2]Shivajoyati, Bangalore, India; [3]McGill University Health Centre, Montreal, QC, Canada; [4]Institute of Experimental and Clinical Therapeutics, University of Guadalajara, Guadalajara, Mexico; [5]Axio Research, Seattle, WA, USA; [6]Janssen Research & Development, LLC, Raritan, NJ, USA; [7]Stanford Center for Clinical Research, Department of Medicine, Stanford University School of Medicine, Stanford, CA, USA; [8]University of Groningen, University Medical Center, Groningen, The Netherlands.

*Presenting author.

INTRODUCTION

- Diabetes is an increasingly large global problem. Between 2017 and 2045, the number of people living with diabetes across the globe is expected to increase from 425 to 629 million[1]
- Canagliflozin (CANA), a sodium glucose co-transporter 2 inhibitor, lowers blood glucose levels by reducing the renal threshold for glucose and increasing urinary glucose excretion[2]
- In the Phase 3 clinical development program, as well as in analyses of specific geographic regions, CANA provided reductions in HbA1c, body weight, and blood pressure (BP) across a broad range of patients[3-6]
- In the CANagliflozin cardioVascular Assessment Study (CANVAS) Program, CANA was associated with a reduced risk of the composite of cardiovascular (CV) death, nonfatal myocardial infarction (MI), or nonfatal stroke compared with placebo (PBO) in patients with type 2 diabetes mellitus (T2DM) and established CV disease or high CV risk[7]
 - The CANVAS Program was a global trial that included participants from 667 sites in 30 countries
 - Several subgroup analyses were prespecified, including in participants by geographic region

OBJECTIVE

- To compare the effects of CANA on CV, renal, and safety outcomes in patients from different geographic regions

METHODS

Study Design and Participants

- The CANVAS Program consisted of 2 multicenter, randomized, double-blind, placebo-controlled trials, CANVAS and CANVAS-R; the design, methods, and primary results of the CANVAS Program have been published
- Eligible participants had T2DM (HbA1c ≥7.0% and ≤10.5%) and were either ≥30 years of age with a history of symptomatic atherosclerotic CV disease or ≥50 years of age with ≥2 risk factors for CV disease
- The prespecified regional groupings were geographical by continent, but with Australia and New Zealand included with Asia (**Figure 1**)

Figure 1. CANVAS Program participants by geographic region.

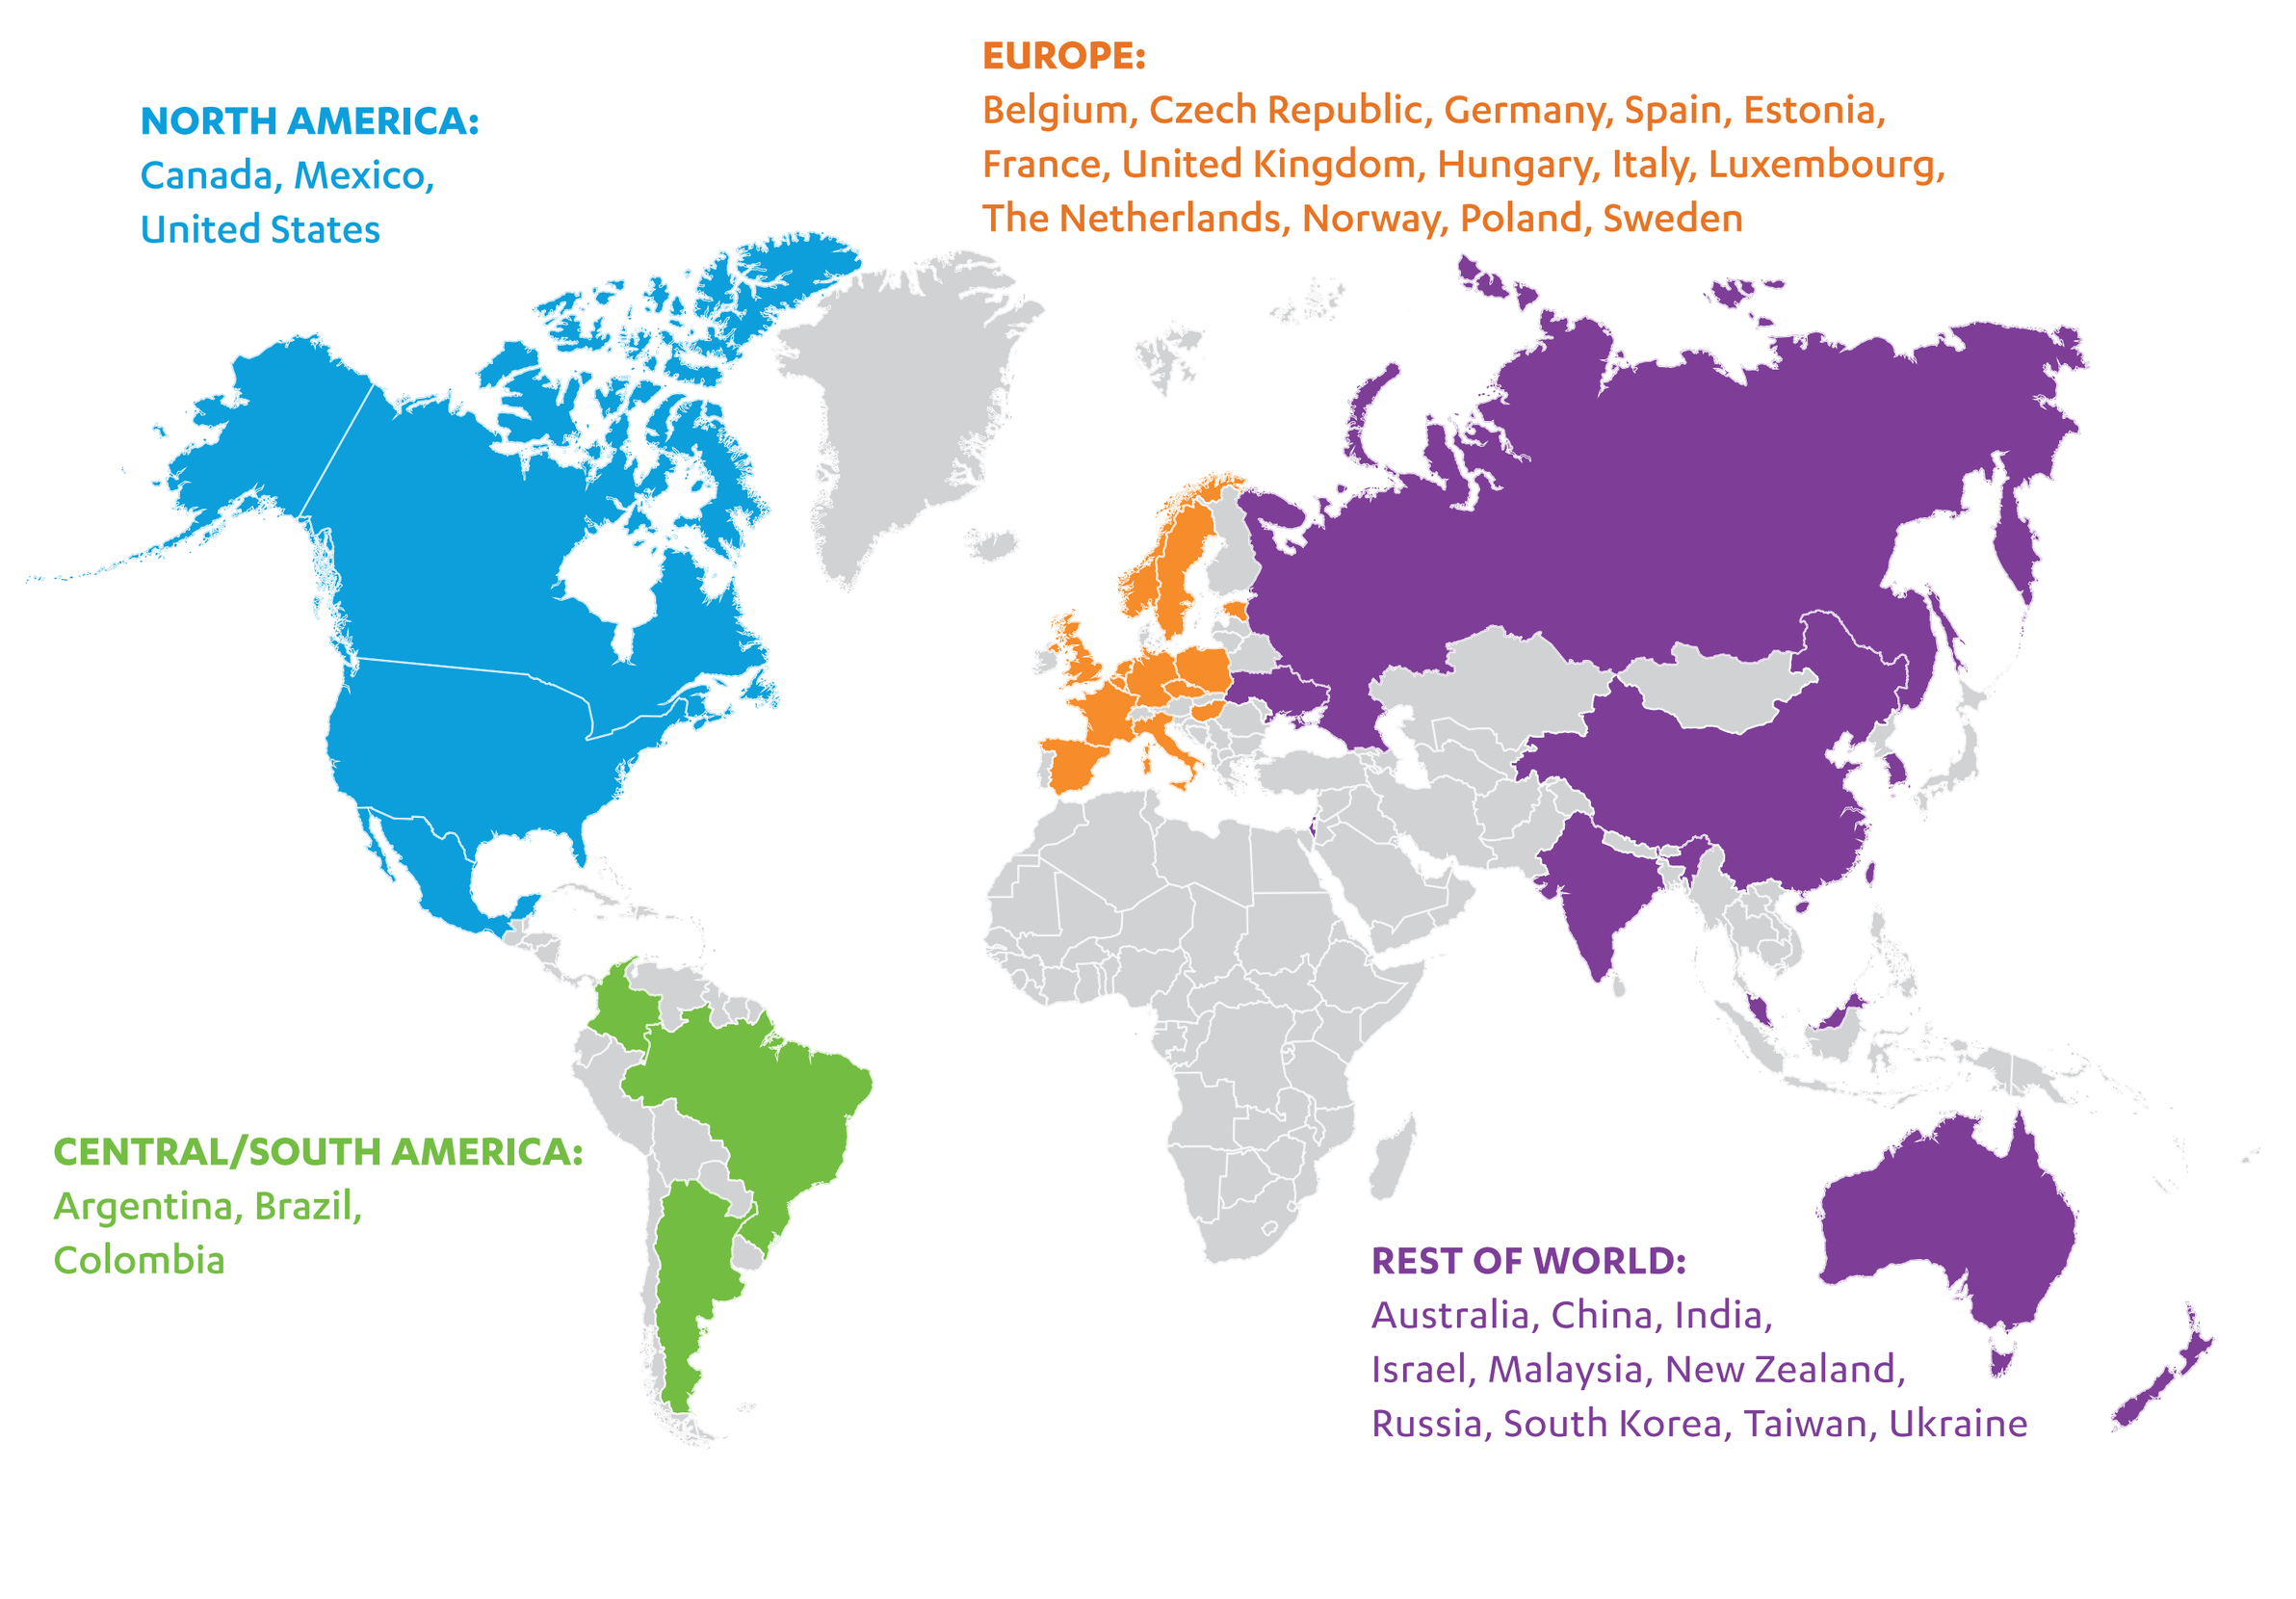
NORTH AMERICA:
Canada, Mexico, United States
EUROPE:
Belgium, Czech Republic, Germany, Spain, Estonia, France, United Kingdom, Hungary, Italy, Luxembourg, The Netherlands, Norway, Poland, Sweden
CENTRAL/SOUTH AMERICA:
Argentina, Brazil, Colombia
REST OF WORLD:
Australia, China, India, Israel, Malaysia, New Zealand, Russia, South Korea, Taiwan, Ukraine

Outcomes

- The effects of CANA on intermediate (HbA1c, body weight, systolic BP, and urinary albumin:creatinine ratio [UACR]), CV, mortality, renal, and safety outcomes were analyzed in participants from different geographic regions
 - All adverse events (AEs) were collected in CANVAS through January 7, 2014
 - After January 7, 2014 in CANVAS and throughout CANVAS-R, AE collection was streamlined; only serious AEs, AEs leading to discontinuation, and all AEs of interest (acute pancreatitis, renal cancer, breast cancer, bladder cancer, photosensitivity, diabetic ketoacidosis, amputation, fracture, venous thromboembolic event, and male genital mycotic infection) were collected

Statistical Analysis

- Unless otherwise specified, efficacy analyses are reported for all CANA doses versus PBO using the integrated CANVAS and CANVAS-R datasets and were based on an intention-to-treat analysis
- The effects of CANA on HbA1c, body weight, and systolic BP were calculated as the mean change across the entire follow-up period using linear mixed models
- The ratio of the geometric mean of postrandomization UACR measures was compared between CANA and PBO
- Hazard ratios (HRs) and 95% confidence intervals (CIs) were estimated using Cox regression models adjusted for study and CV risk subgroups, with treatment included as a fixed covariate
- Annualized incidence rates were calculated per 1000 patient-years of follow-up
- *P* values for heterogeneity were calculated by including treatment-by-subgroup interactions in the Cox regression models and are provided for descriptive purposes without adjustment for multiplicity

RESULTS

Participants

- Overall, 2430 participants (24.0%) were from North America, 1021 (10.1%) were from Central/South America, 3609 (35.6%) were from Europe, and 3082 (30.4%) were from the rest of the world (**Table 1**)

Effects on Intermediate Outcomes

- Compared with PBO, CANA reduced HbA1c, body weight, systolic BP, and UACR in participants from different regions (**Figure 2**)
- Heterogeneity was observed in the effects of CANA on body weight in participants from different regions of the world. As expected, greater reductions in body weight were seen in participants from the geographic region subgroups with higher baseline body mass index (BMI; *P* heterogeneity <0.0001; **Figure 2B**)

Effects on CV, Mortality, and Renal Outcomes

- The effect of CANA on CV and renal outcomes appeared to be consistent in participants from different geographic regions (**Figure 3**)

Safety

- The safety profile of CANA was generally similar in participants from different geographic regions (**Figure 4**)

LIMITATIONS

- The CANVAS Program was not powered to detect differences between geographic subgroups, and interpretation of subgroup data can be challenging due to nonrandomized groups and small sample sizes
- Interpretation of these analyses is limited by the large number of subgroup comparisons

SUMMARY

- **CANA appears to provide consistent benefits in intermediate, CV, mortality, and renal outcomes in participants across different geographic regions**
- **The safety of CANA was generally similar in participants from different geographic regions**

Figure 2. PBO-subtracted differences (95% CI) in intermediate outcomes across the entire follow-up period with CANA in participants from different geographic regions.

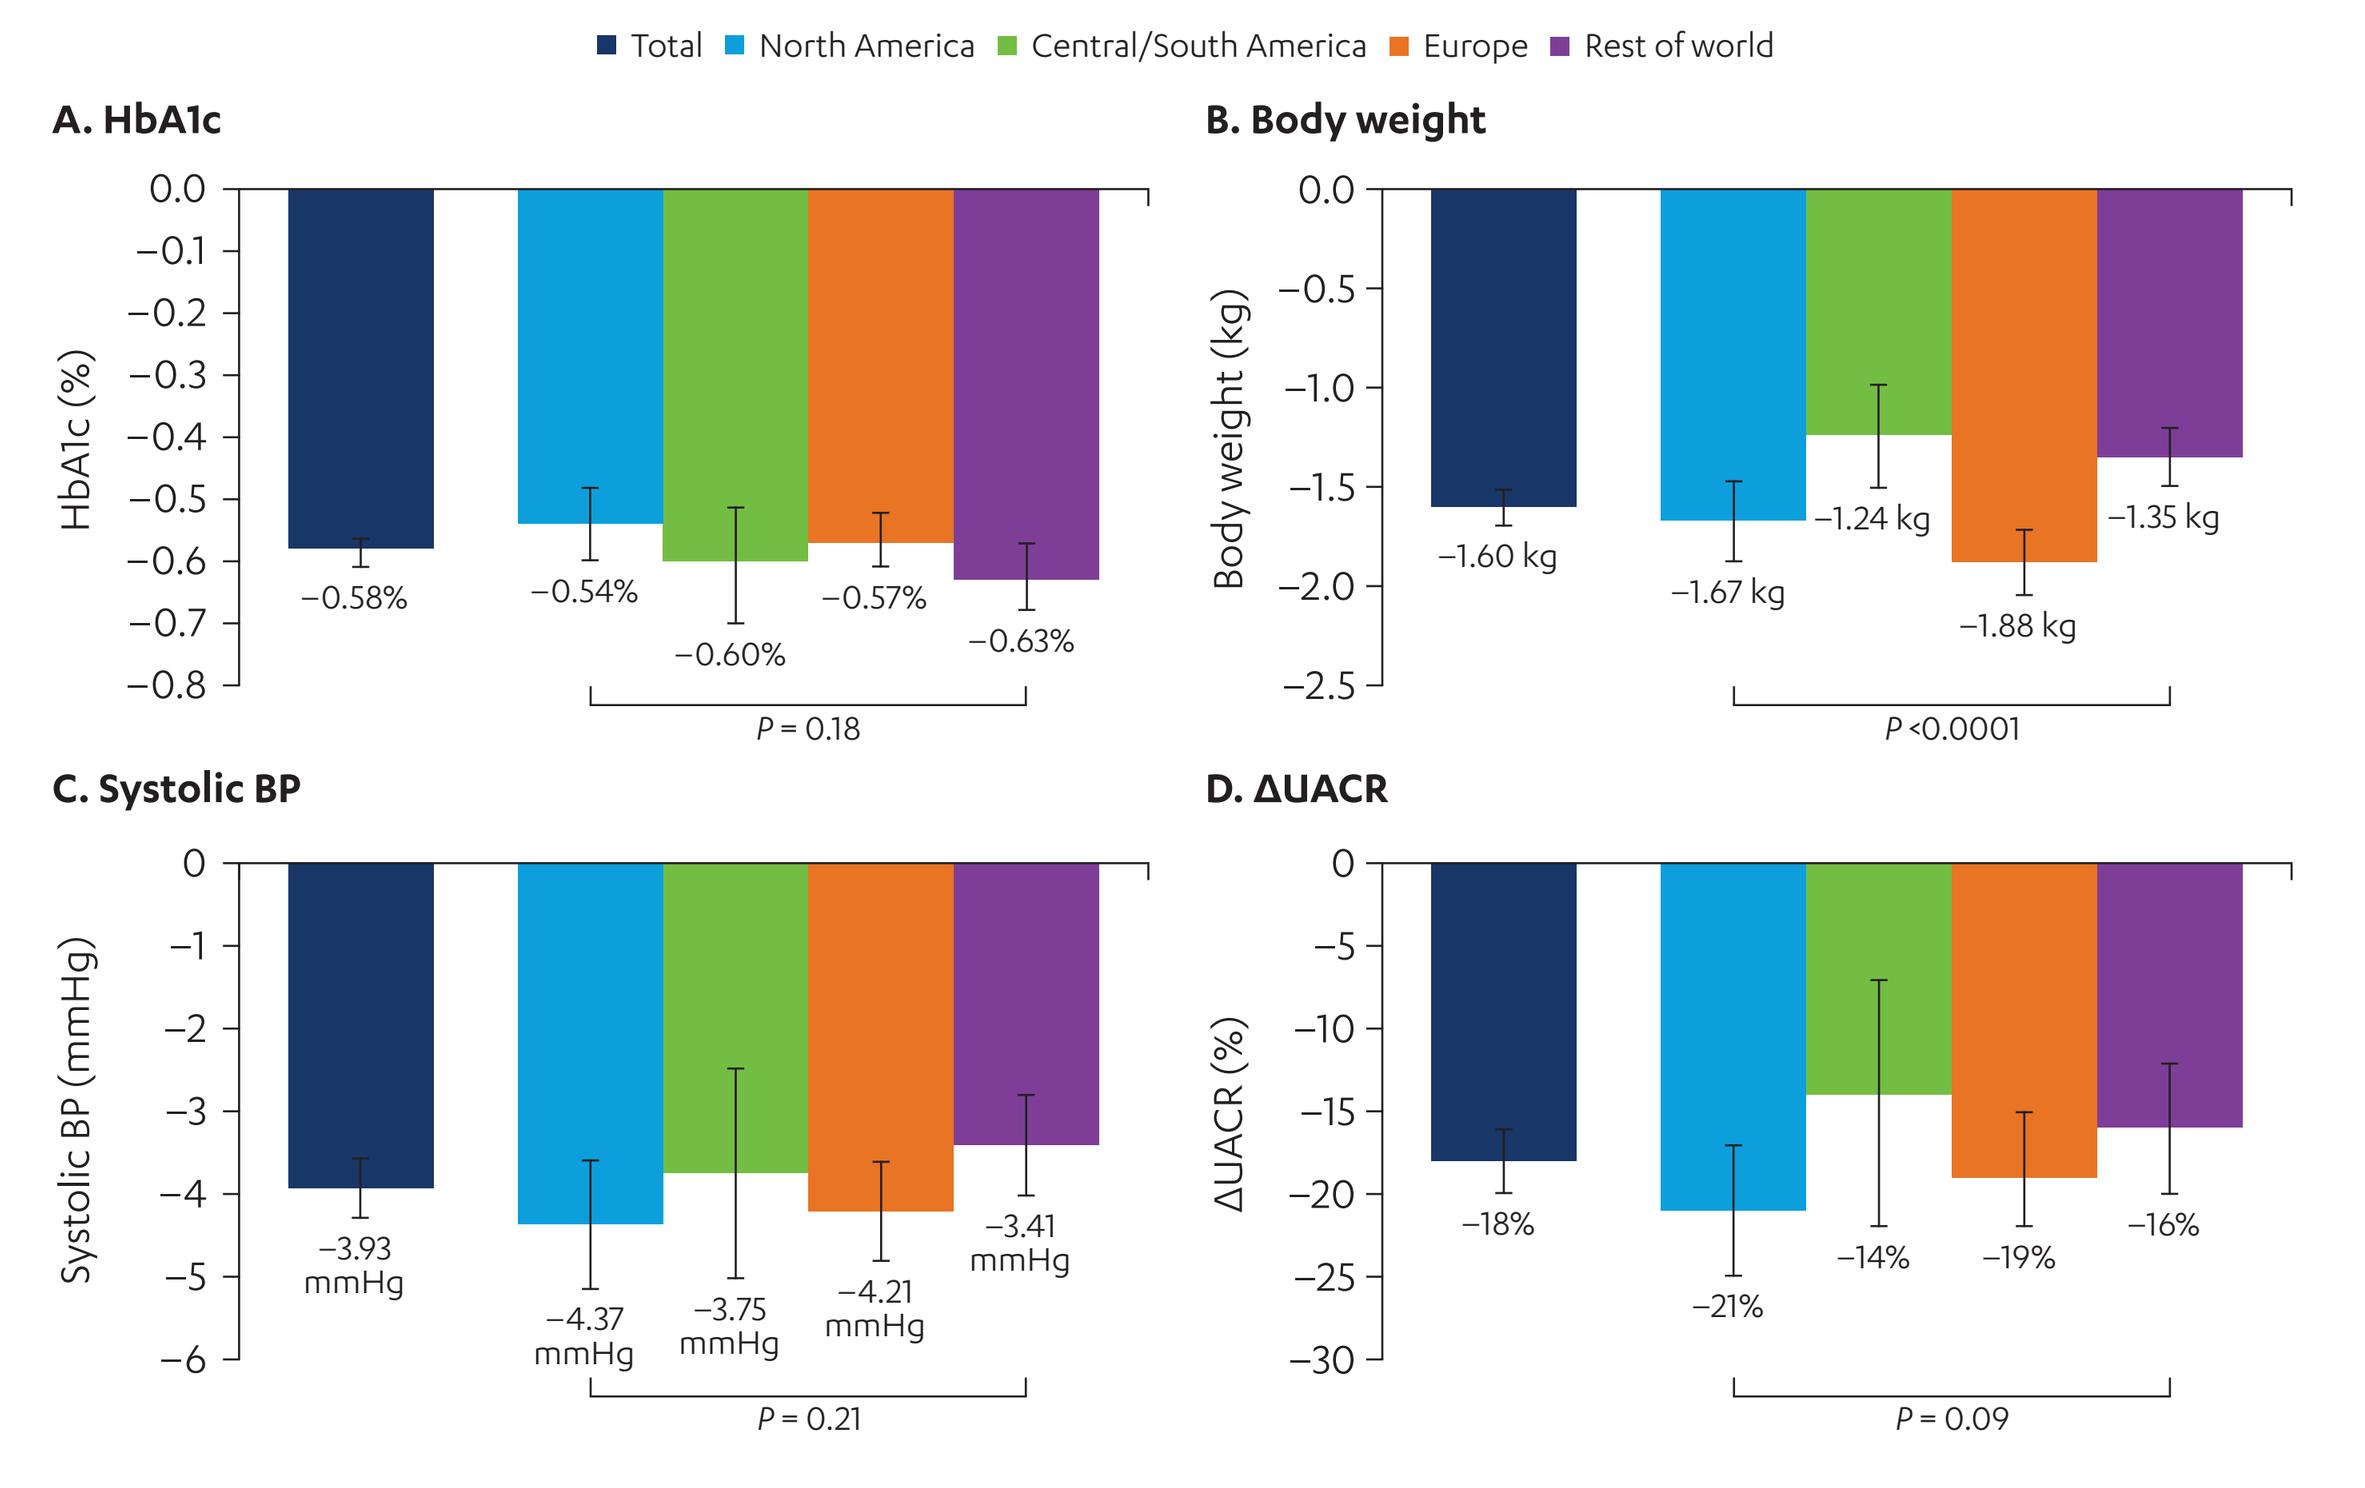
Total
North America
Central/South America
Europe
Rest of world
A. HbA1c
HbA1c (%)
−0.58%
−0.54%
−0.60%
−0.57%
−0.63%
P = 0.18
B. Body weight
Body weight (kg)
−1.60 kg
−1.67 kg
−1.24 kg
−1.88 kg
−1.35 kg
P <0.0001
C. Systolic BP
Systolic BP (mmHg)
−3.93 mmHg
−4.37 mmHg
−3.75 mmHg
−4.21 mmHg
−3.41 mmHg
P = 0.21
D. ΔUACR
ΔUACR (%)
−18%
−21%
−14%
−19%
−16%
P = 0.09

Table 1. Baseline Characteristics for the CANVAS Program by Geographic Region*

	North America (n = 2430)	Central/ South America (n = 1021)	Europe (n = 3609)	Rest of world (n = 3082)	*P* value
Age, y	63.2 (8.4)	63.2 (8.9)	64.8 (7.8)	61.7 (8.0)	<0.001
Female, n (%)	776 (31.9)	480 (47.0)	1097 (30.4)	1280 (41.5)	<0.001
Race, n (%)†					
White	1955 (80.5)	789 (77.3)	3389 (93.9)	1811 (58.8)	<0.001
Asian	58 (2.4)	11 (1.1)	19 (0.5)	1196 (38.8)	
Black or African American	197 (8.1)	127 (12.4)	10 (0.3)	2 (0.1)	
Other	220 (9.1)	94 (9.2)	191 (5.3)	73 (2.4)	
BMI, kg/m^2	33.3 (6.7)	30.7 (5.3)	32.6 (5.4)	30.6 (5.7)	<0.001
HbA1c, %	8.2 (1.0)	8.4 (1.0)	8.1 (0.9)	8.4 (1.0)	<0.001
Systolic BP, mmHg	133.8 (16.4)	132.9 (16.5)	140.1 (15.7)	136.1 (14.2)	<0.001
Duration of diabetes, y	14.5 (7.9)	13.2 (8.2)	14.3 (7.8)	12.1 (7.2)	<0.001
CV disease history, n (%)‡	1558 (64.1)	728 (71.3)	2335 (64.7)	2035 (66.0)	<0.001
eGFR, mL/min/1.73 m^2	75.5 (21.1)	80.0 (23.6)	75.3 (19.8)	77.5 (19.5)	<0.001
UACR, mg/g, median (IQR)	11.8 (6.3-40.6)	11.2 (6.7-40.4)	12.4 (6.6-45.6)	12.7 (6.9-40.5)	0.18
AHA use, n (%)					
Insulin	1230 (50.6)	483 (47.3)	2083 (57.7)	1299 (42.1)	<0.001
Sulfonylurea	1045 (43.0)	439 (43.0)	1113 (30.8)	1764 (57.2)	<0.001
Metformin	1807 (74.4)	839 (82.2)	2827 (78.3)	2352 (76.3)	<0.001
GLP-1 receptor agonist	109 (4.5)	6 (0.6)	272 (7.5)	20 (0.6)	<0.001
DPP-4 inhibitor	345 (14.2)	106 (10.4)	542 (15.0)	268 (8.7)	<0.001
CV medication use, n (%)					
RAAS inhibitor use	1909 (78.6)	808 (79.1)	2956 (81.9)	2443 (79.3)	0.005
Statin	1890 (77.8)	688 (67.4)	2971 (82.3)	2051 (66.5)	<0.001
Antithrombotic	1761 (72.5)	666 (65.2)	2741 (75.9)	2303 (74.7)	<0.001
Beta blocker	1182 (48.6)	488 (47.8)	2188 (60.6)	1563 (50.7)	<0.001
Diuretic	1108 (45.6)	356 (34.9)	1876 (52.0)	1150 (37.3)	<0.001

eGFR, estimated glomerular filtration rate; IQR, interquartile range; AHA, antihyperglycemic agent; GLP-1, glucagon-like peptide-1; DPP-4, dipeptidyl peptidase-4; RAAS, renin-angiotensin-aldosterone system; SD, standard deviation.
*Data are mean (SD) unless otherwise noted.
†Percentages may not total 100.0% due to rounding.
‡As defined in the protocol.

Figure 3. Effect of CANA on CV, mortality, and renal outcomes in participants from different geographic regions.

		Number of patients with an event	Patients with an event per 1000 patient-years: CANA	PBO	HR (95% CI)	Interaction *P* value
CV death, nonfatal MI, or nonfatal stroke*	Total	1011	26.9	31.5	0.86 (0.75, 0.97)	0.89
	North America	252	26.7	30.3	0.84 (0.65, 1.09)	
	Central/South America	73	26.7	33.8	0.84 (0.53, 1.33)	
	Europe	370	28.1	35.0	0.80 (0.65, 0.99)	
	Rest of world	316	26.0	28.2	0.94 (0.75, 1.18)	
CV death	Total	453	11.6	12.8	0.87 (0.72, 1.06)	0.75
	North America	117	11.9	11.3	1.00 (0.68, 1.46)	
	Central/South America	44	17.1	17.8	1.02 (0.56, 1.85)	
	Europe	142	10.5	12.3	0.79 (0.57, 1.11)	
	Rest of world	150	11.4	13.5	0.84 (0.60, 1.16)	
Nonfatal MI	Total	374	9.7	11.6	0.85 (0.69, 1.05)	0.24
	North America	96	9.0	13.2	0.65 (0.43, 0.98)	
	Central/South America	16	3.7	10.0	0.40 (0.14, 1.16)	
	Europe	161	12.5	14.2	0.90 (0.65, 1.24)	
	Rest of world	101	8.6	8.1	1.09 (0.72, 1.64)	
Nonfatal stroke	Total	274	7.1	8.4	0.90 (0.71, 1.15)	0.22
	North America	58	6.0	6.8	0.90 (0.53, 1.54)	
	Central/South America	16	7.4	5.4	1.46 (0.53, 4.05)	
	Europe	106	7.0	11.2	0.64 (0.43, 0.94)	
	Rest of world	94	8.1	7.4	1.22 (0.80, 1.87)	
All-cause mortality	Total	681	17.3	19.5	0.87 (0.74, 1.01)	0.86
	North America	194	19.2	19.6	0.94 (0.70, 1.26)	
	Central/South America	64	24.3	26.7	0.98 (0.60, 1.61)	
	Europe	226	16.8	19.4	0.81 (0.62, 1.06)	
	Rest of world	197	15.0	17.8	0.83 (0.62, 1.11)	
Hospitalization for heart failure	Total	243	5.5	8.7	0.67 (0.52, 0.87)	0.53
	North America	54	5.5	6.5	0.81 (0.47, 1.41)	
	Central/South America	17	4.4	10.0	0.45 (0.16, 1.22)	
	Europe	127	8.5	13.0	0.72 (0.51, 1.03)	
	Rest of world	45	2.7	5.4	0.51 (0.28, 0.92)	
40% reduction in eGFR, ESKD, or renal death	Total	249	5.5	9.0	0.60 (0.47, 0.77)	0.32
	North America	68	5.3	11.0	0.47 (0.29, 0.76)	
	Central/South America	13	2.9	8.1	0.35 (0.11, 1.16)	
	Europe	92	6.0	9.6	0.60 (0.40, 0.91)	
	Rest of world	76	5.7	7.2	0.81 (0.51, 1.28)	
dSCr, ESKD, or renal death	Total	73	1.5	2.8	0.53 (0.33, 0.84)	0.67
	North America	25	1.7	4.4	0.37 (0.16, 0.83)	
	Central/South America	2	0	1.8	–†	
	Europe	30	1.9	3.1	0.60 (0.29, 1.24)	
	Rest of world	16	1.2	1.5	0.85 (0.31, 2.32)	
Progression of albuminuria	Total	2455	89.4	128.7	0.73 (0.67, 0.79)	0.76
	North America	589	87.0	125.4	0.74 (0.63, 0.87)	
	Central/South America	224	110.3	151.5	0.77 (0.59, 1.00)	
	Europe	838	86.3	130.0	0.68 (0.59, 0.78)	
	Rest of world	804	90.4	123.6	0.77 (0.67, 0.88)	

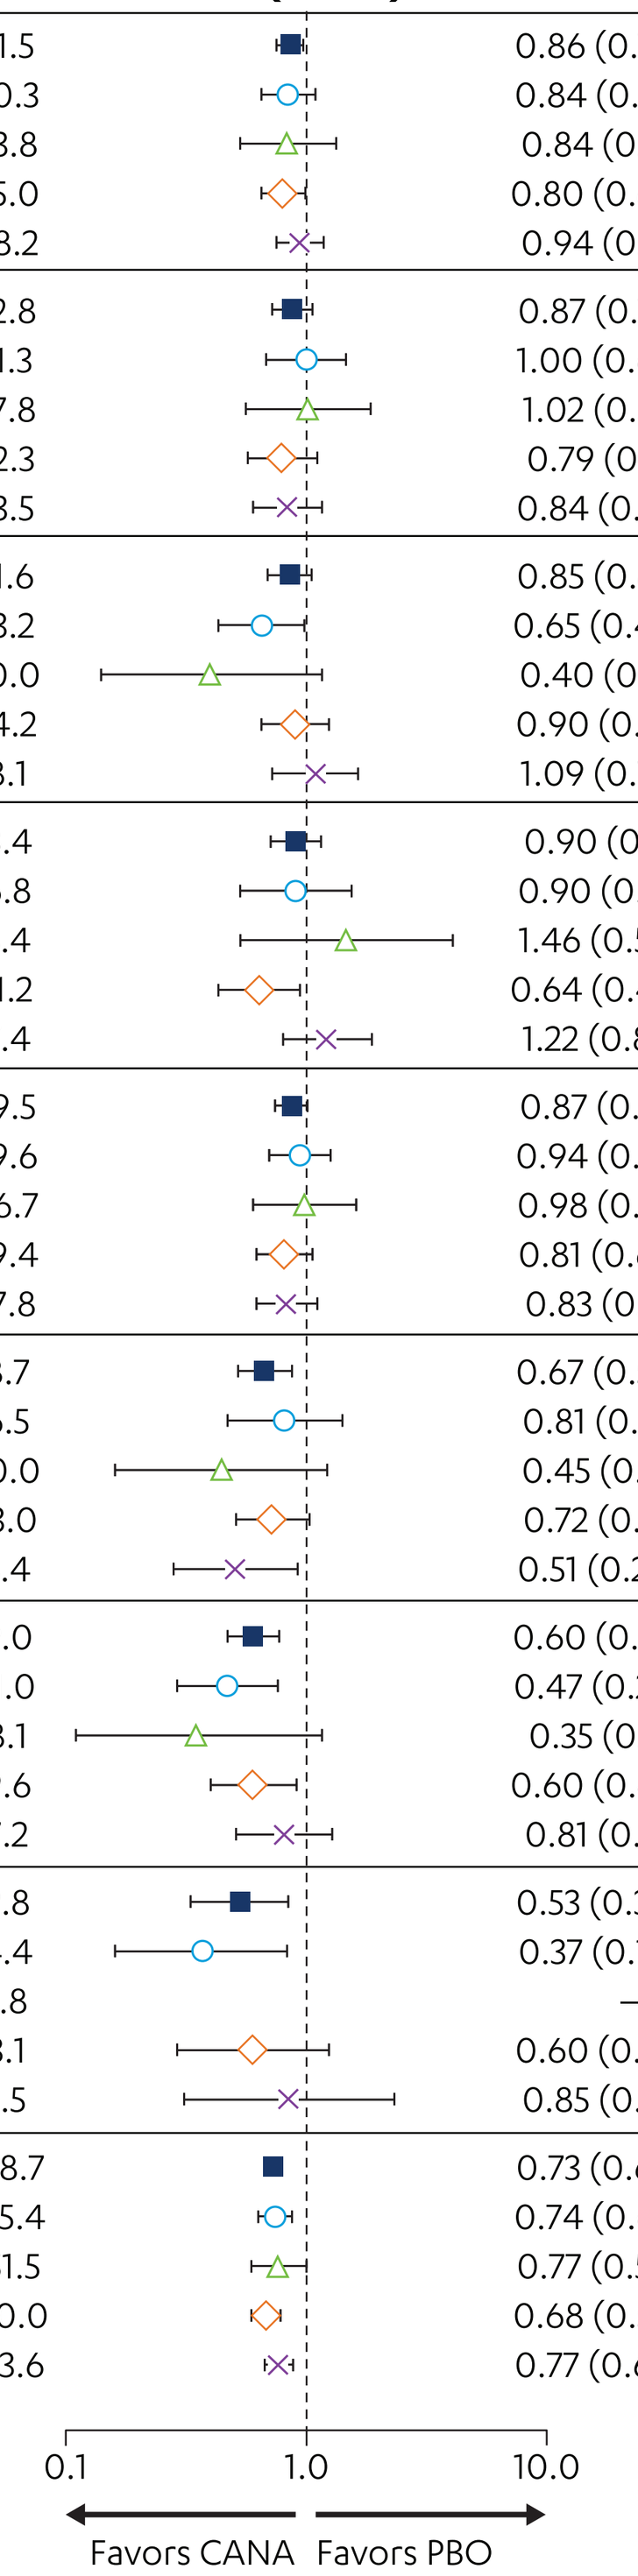
0.1
1.0
10.0
Favors CANA
Favors PBO

ESKD, end-stage kidney disease; dSCr, doubling of serum creatinine.
*Primary outcome.
†HRs and 95% CIs were calculated for all subgroups with >10 events.

Figure 4. Effect of CANA on safety outcomes in participants from different geographic regions.*

		Number of patients with an event	Patients with an event per 1000 patient-years: CANA	PBO	HR (95% CI)	Interaction *P* value
Serious AEs	Total	3280	104.3	120.0	0.93 (0.87, 1.00)	0.39
	North America	779	100.3	113.6	0.97 (0.84, 1.12)	
	Central/South America	226	101.3	101.5	1.03 (0.79, 1.34)	
	Europe	1299	122.1	139.8	0.94 (0.84, 1.06)	
	Rest of world	976	90.2	109.0	0.86 (0.75, 0.97)	
AEs leading to discontinuation	Total	1025	35.5	32.8	1.13 (0.99, 1.28)	0.04
	North America	270	33.4	41.8	0.84 (0.65, 1.07)	
	Central/South America	66	30.2	28.9	1.09 (0.67, 1.78)	
	Europe	441	46.5	39.4	1.22 (1.00, 1.49)	
	Rest of world	248	27.2	20.8	1.32 (1.01, 1.73)	
Serious and nonserious AEs of interest collected in the CANVAS Program						
Diabetic ketoacidosis	Total	18	0.6	0.3	2.33 (0.76, 7.17)	–
	North America	6	0.8	0.3	–†	
	Central/South America	1	0.7	0	–†	
	Europe	7	0.6	0.4	–†	
	Rest of world	4	0.4	0.2	–†	
Amputation	Total	187	6.3	3.4	1.97 (1.41, 2.75)	0.30
	North America	49	5.7	4.7	1.22 (0.67, 2.24)	
	Central/South America	21	12.6	3.6	3.74 (1.25, 11.2)	
	Europe	70	7.0	3.5	2.04 (1.17, 3.56)	
	Rest of world	47	5.0	2.2	2.33 (1.15, 4.71)	
Fracture	Total	496	15.4	11.9	1.26 (1.04, 1.52)	0.23
	North America	143	15.7	16.4	0.92 (0.66, 1.30)	
	Central/South America	30	15.1	9.1	1.57 (0.73, 3.36)	
	Europe	200	18.7	13.4	1.37 (1.01, 1.86)	
	Rest of world	123	12.0	7.9	1.46 (0.99, 2.17)	
Male genital mycotic infection	Total	456	31.7	9.6	3.76 (2.91, 4.86)	0.12
	North America	98	24.4	7.7	3.56 (2.01, 6.29)	
	Central/South America	43	57.5	7.3	8.31 (2.97, 23.3)	
	Europe	245	48.5	13.4	4.28 (3.01, 6.09)	
	Rest of world	70	14.6	7.0	2.12 (1.19, 3.75)	
Serious and nonserious AEs of interest collected in CANVAS alone‡						
Osmotic diuresis	Total	312	34.5	13.3	2.80 (2.06, 3.81)	0.47
	North America	106	42.1	12.2	4.00 (2.19, 7.29)	
	Central/South America	7	26.3	0	–†	
	Europe	123	42.8	19.9	2.30 (1.44, 3.67)	
	Rest of world	76	22.0	10.5	2.17 (1.23, 3.82)	
Volume depletion	Total	266	26.0	18.5	1.44 (1.09, 1.90)	0.16
	North America	124	38.9	37.6	1.08 (0.73, 1.59)	
	Central/South America	14	45.0	13.2	3.54 (0.79, 15.8)	
	Europe	83	26.4	19.0	1.42 (0.86, 2.35)	
	Rest of world	45	13.4	5.6	2.43 (1.13, 5.22)	
Hypoglycemia	Total	551	50.0	46.4	1.13 (0.94, 1.35)	0.81
	North America	208	64.9	63.9	1.11 (0.83, 1.49)	
	Central/South America	32	78.8	72.6	1.15 (0.55, 2.38)	
	Europe	131	37.8	38.9	0.99 (0.69, 1.44)	
	Rest of world	180	45.8	37.2	1.27 (0.92, 1.74)	
Urinary tract infection	Total	443	40.4	37.0	1.11 (0.90, 1.36)	0.84
	North America	140	44.8	40.6	1.15 (0.80, 1.86)	
	Central/South America	21	48.8	52.8	0.90 (0.37, 2.17)	
	Europe	159	46.2	46.5	0.99 (0.71, 1.39)	
	Rest of world	123	31.0	26.0	1.21 (0.83, 1.79)	
Female genital mycotic infection	Total	196	68.8	17.5	4.37 (2.78, 6.88)	0.65
	North America	80	115.1	29.5	4.92 (2.37, 10.2)	
	Central/South America	9	60.4	37.8	–†	
	Europe	60	77.7	16.2	5.15 (2.06, 12.9)	
	Rest of world	47	37.5	10.6	3.75 (1.59, 8.83)	
Renal-related AEs (including acute kidney injury)	Total	214	19.7	17.4	1.16 (0.86, 1.56)	0.29
	North America	80	23.3	28.4	0.85 (0.54, 1.34)	
	Central/South America	8	26.3	6.6	–†	
	Europe	45	13.9	11.4	1.22 (0.63, 2.36)	
	Rest of world	81	21.3	15.4	1.41 (0.87, 2.31)	

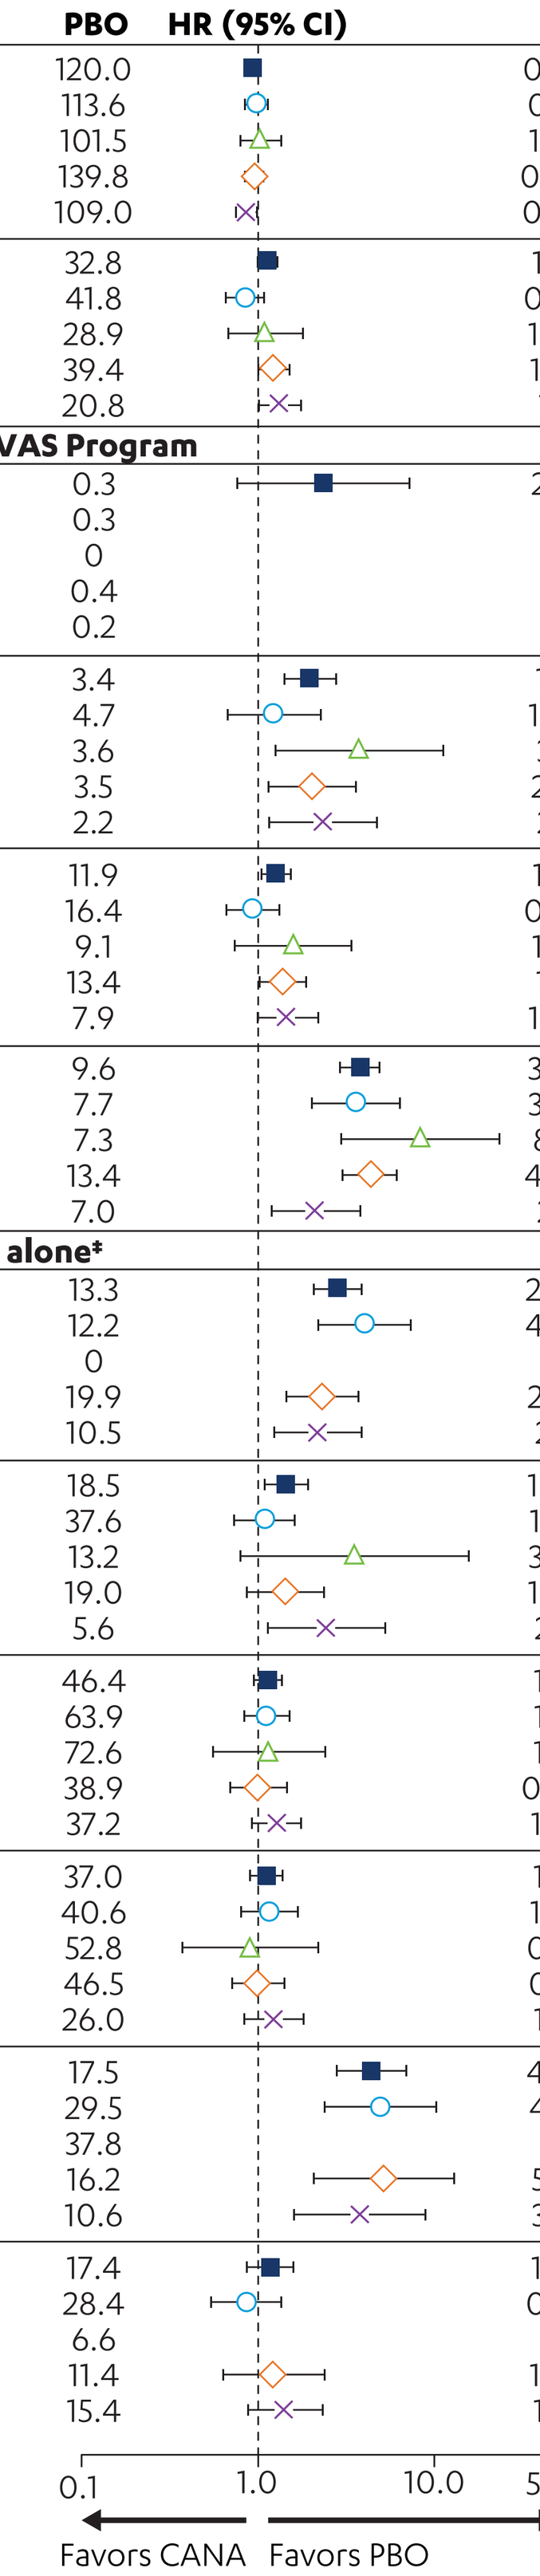
0.1
1.0
10.0
50.0
Favors CANA
Favors PBO

*Analyses were performed on data from the on-treatment data set (on-treatment or 30 days after discontinuation), except for fracture, amputation, and diabetic ketoacidosis outcomes, which included all events at any time point in all patients who underwent randomization and received ≥1 dose of CANA or PBO.
†HRs and 95% CIs were calculated for all subgroups with >10 events.
‡For these AEs, the annualized incidence rates are reported with data from CANVAS alone through January 7, 2014 because, after this time, only serious AEs or AEs leading to discontinuation were collected. In CANVAS-R, only serious AEs or AEs leading to discontinuation were collected. Owing to the differences between the 2 trials in methods of collection of the data, an integrated analysis of these AEs is not possible.

REFERENCES

1. International Diabetes Federation. IDF Diabetes Atlas, Eighth Edition. International Diabetes Federation; 2017. www.diabetesatlas.org. Accessed January 16, 2018.
2. Rosenthal N, et al. *Ann N Y Acad Sci*. 2015;1358:28-43.
3. John M, et al. *Int J Clin Pract*. 2016;70(9):775-785.
4. Lavalle-González FJ, et al. *Curr Med Res Opin*. 2016;32(3):427-439.
5. Prasanna Kumar KM, et al. *Indian J Endocrinol Metab*. 2016;20(3):372-380.
6. Ji L, et al. *Diabetes Obes Metab*. 2015;17(1):23-31.
7. Neal B, et al. *N Engl J Med*. 2017;377(7):644-657.

ACKNOWLEDGMENTS

This study was supported by Janssen Research & Development, LLC. Medical writing support was provided by Dana Tabor, PhD, of MedErgy, and was funded by Janssen Global Services, LLC.

Canagliflozin has been developed by Janssen Research & Development, LLC, in collaboration with Mitsubishi Tanabe Pharma Corporation.

CONTACT INFORMATION

Carol Wysham
Washington State University
400 East Fifth Ave
Spokane, WA 99202
Email: CWysham@rockwoodclinic.com

POSTER PRESENTED AT THE 78TH SCIENTIFIC SESSIONS OF THE AMERICAN DIABETES ASSOCIATION (ADA); JUNE 22-26, 2018; ORLANDO, FLORIDA.